From Survivor Story to Public Health Impact: Families Fighting Flu's Novel Approach to Influenza Prevention

KADEN BLAZE

FAMILIES FIGHTING FLU™

Created by FFF, this comic reimagines Kaden as "Kaden Blaze," a superhero who helps audiences of all ages understand the serious impact of influenza and the importance of prevention. By using an engaging, accessible storytelling format, we:

- **Reached impressionable young audiences**
- **Reached diverse audiences in both English and Spanish**
- **Expanded the impact of traditional health communication**

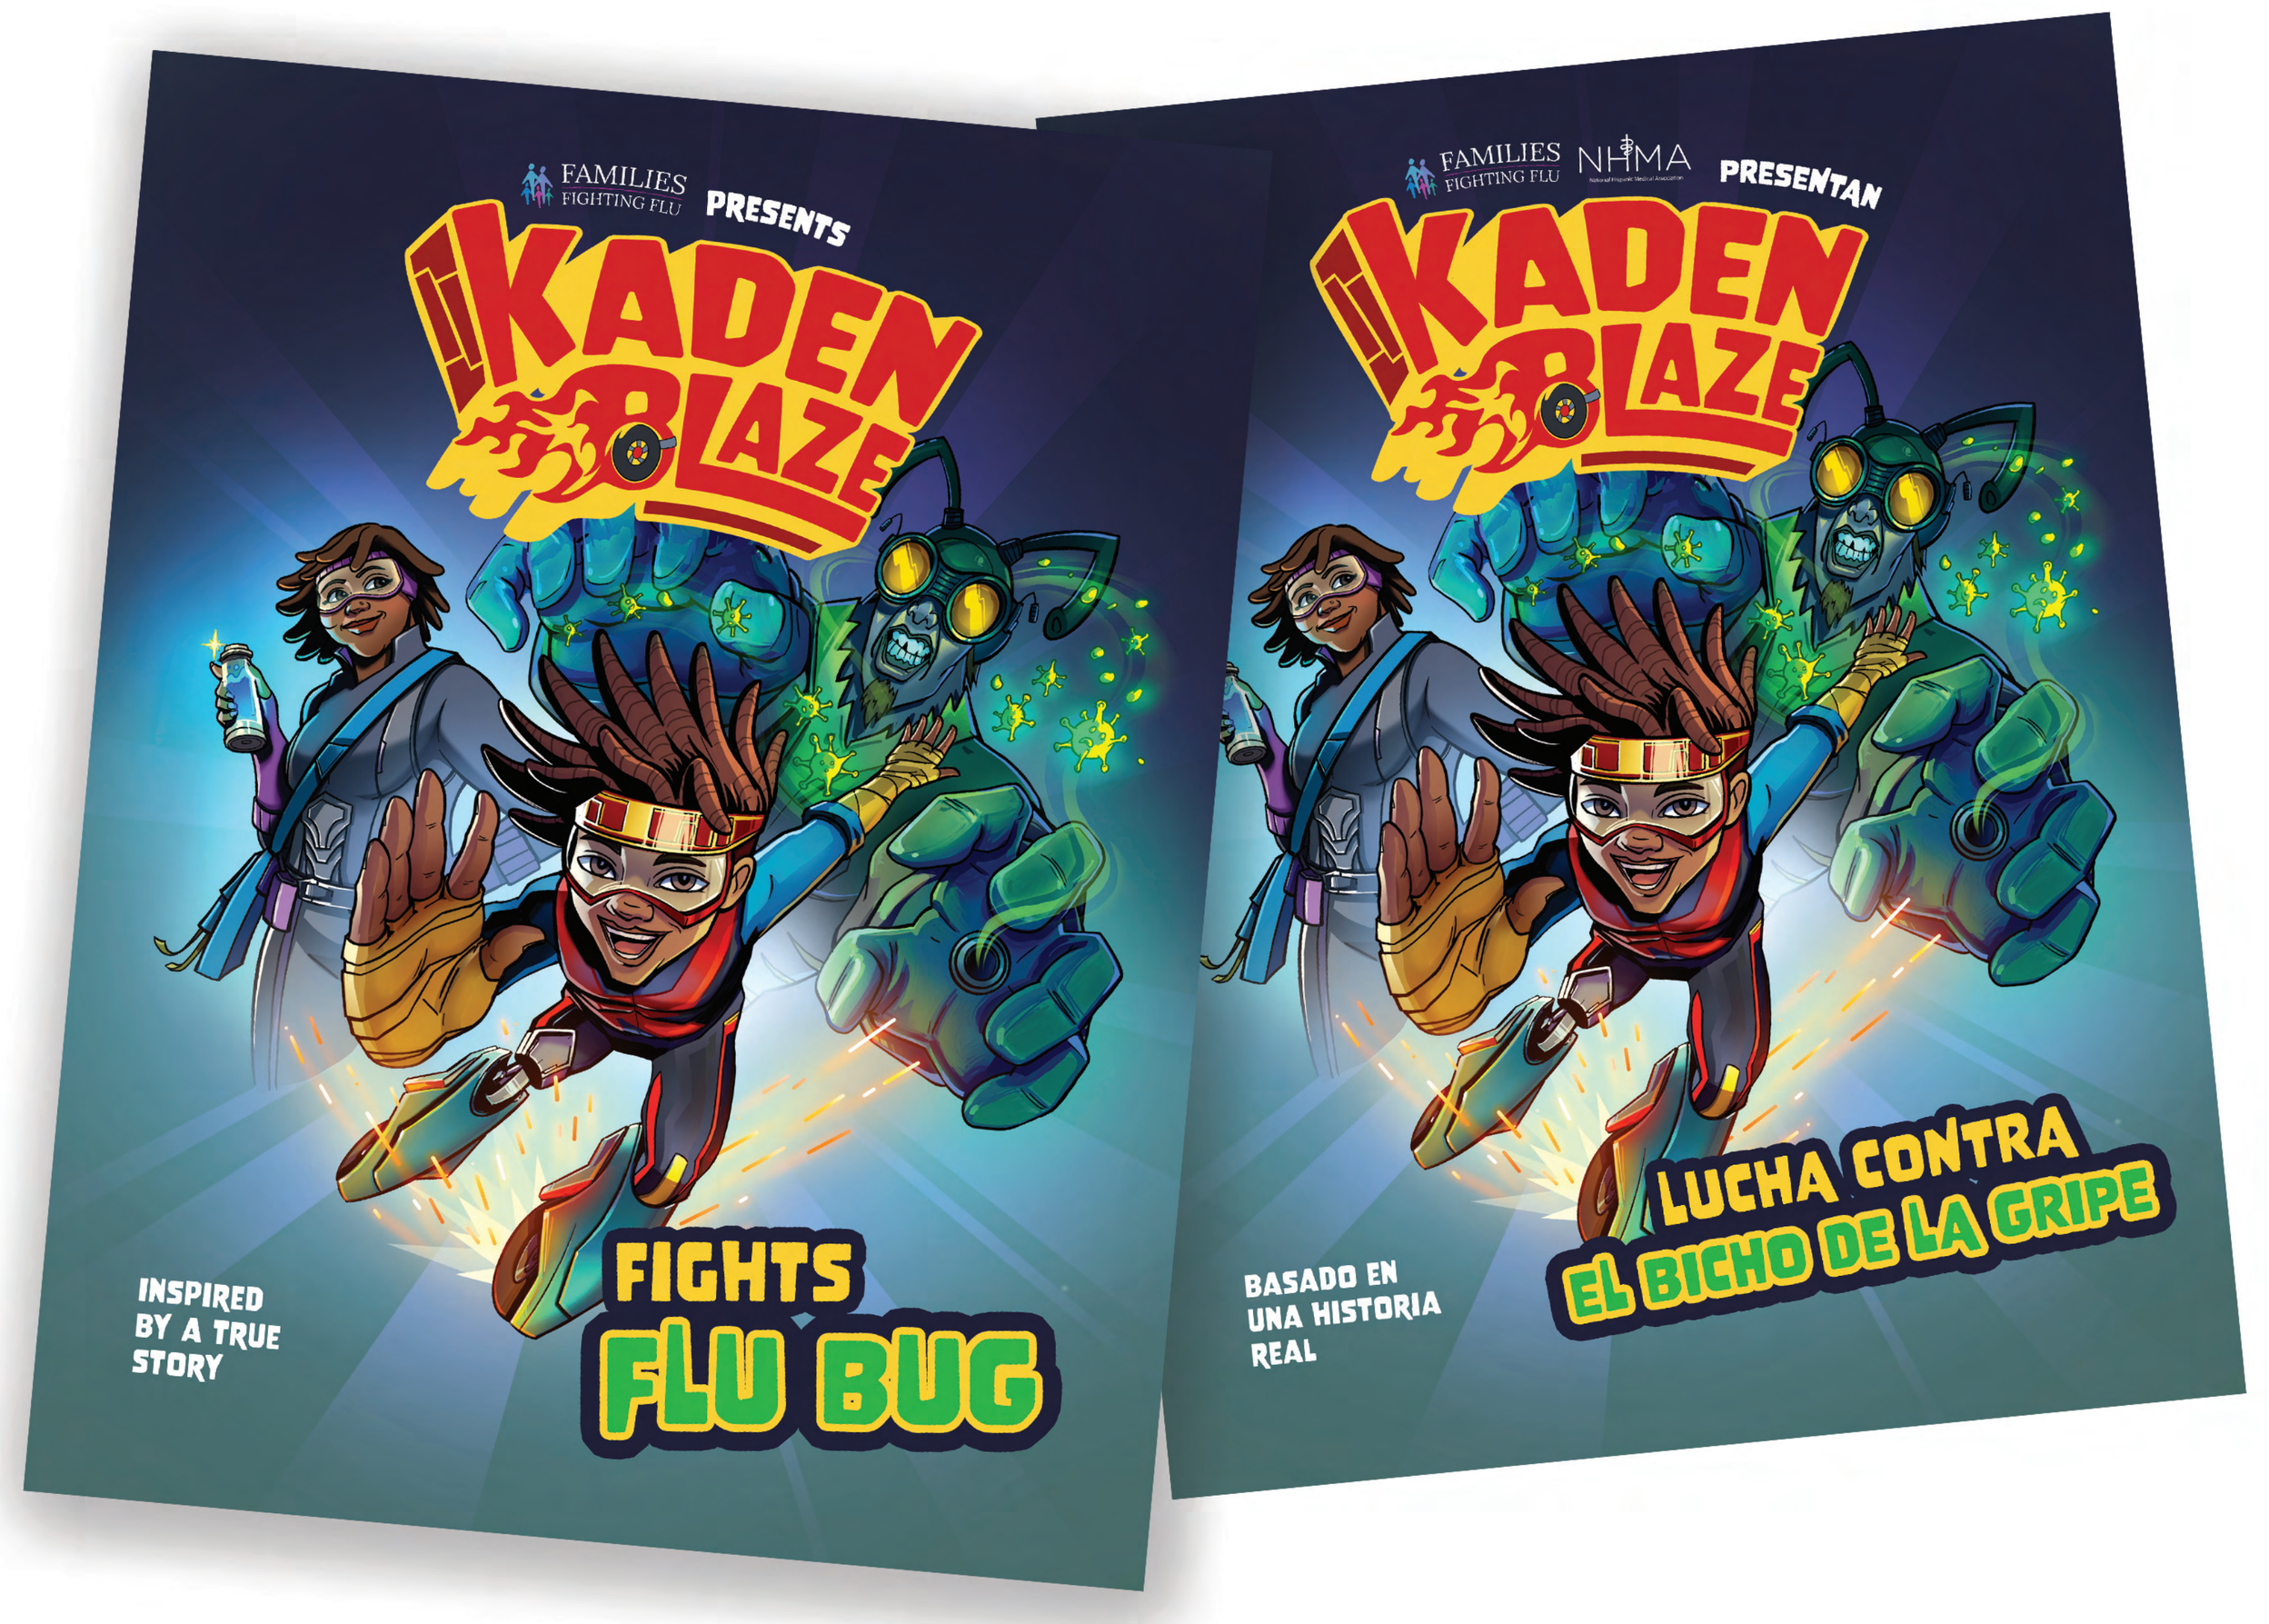

FAMILIES FIGHTING FLU PRESENTS
KADEN BLAZE
INSPIRED BY A TRUE STORY
FIGHTS FLU BUG
FAMILIES FIGHTING FLU NHMA PRESENTAN
KADEN BLAZE
BASADO EN UNA HISTORIA REAL
LUCHA CONTRA EL BICHO DE LA GRIPE

Kaden Stevenson's journey is one of the many powerful family stories shared through Families Fighting Flu (FFF). After getting the flu in 2022, which led to multi-organ failure and the loss of both legs, Kaden's experience became a catalyst for advocacy. Through FFF, his **REAL STORY** was transformed into a new educational resource: the comic book

Kaden Blaze Fights the Flu Bug.

RESOURCES HERE!

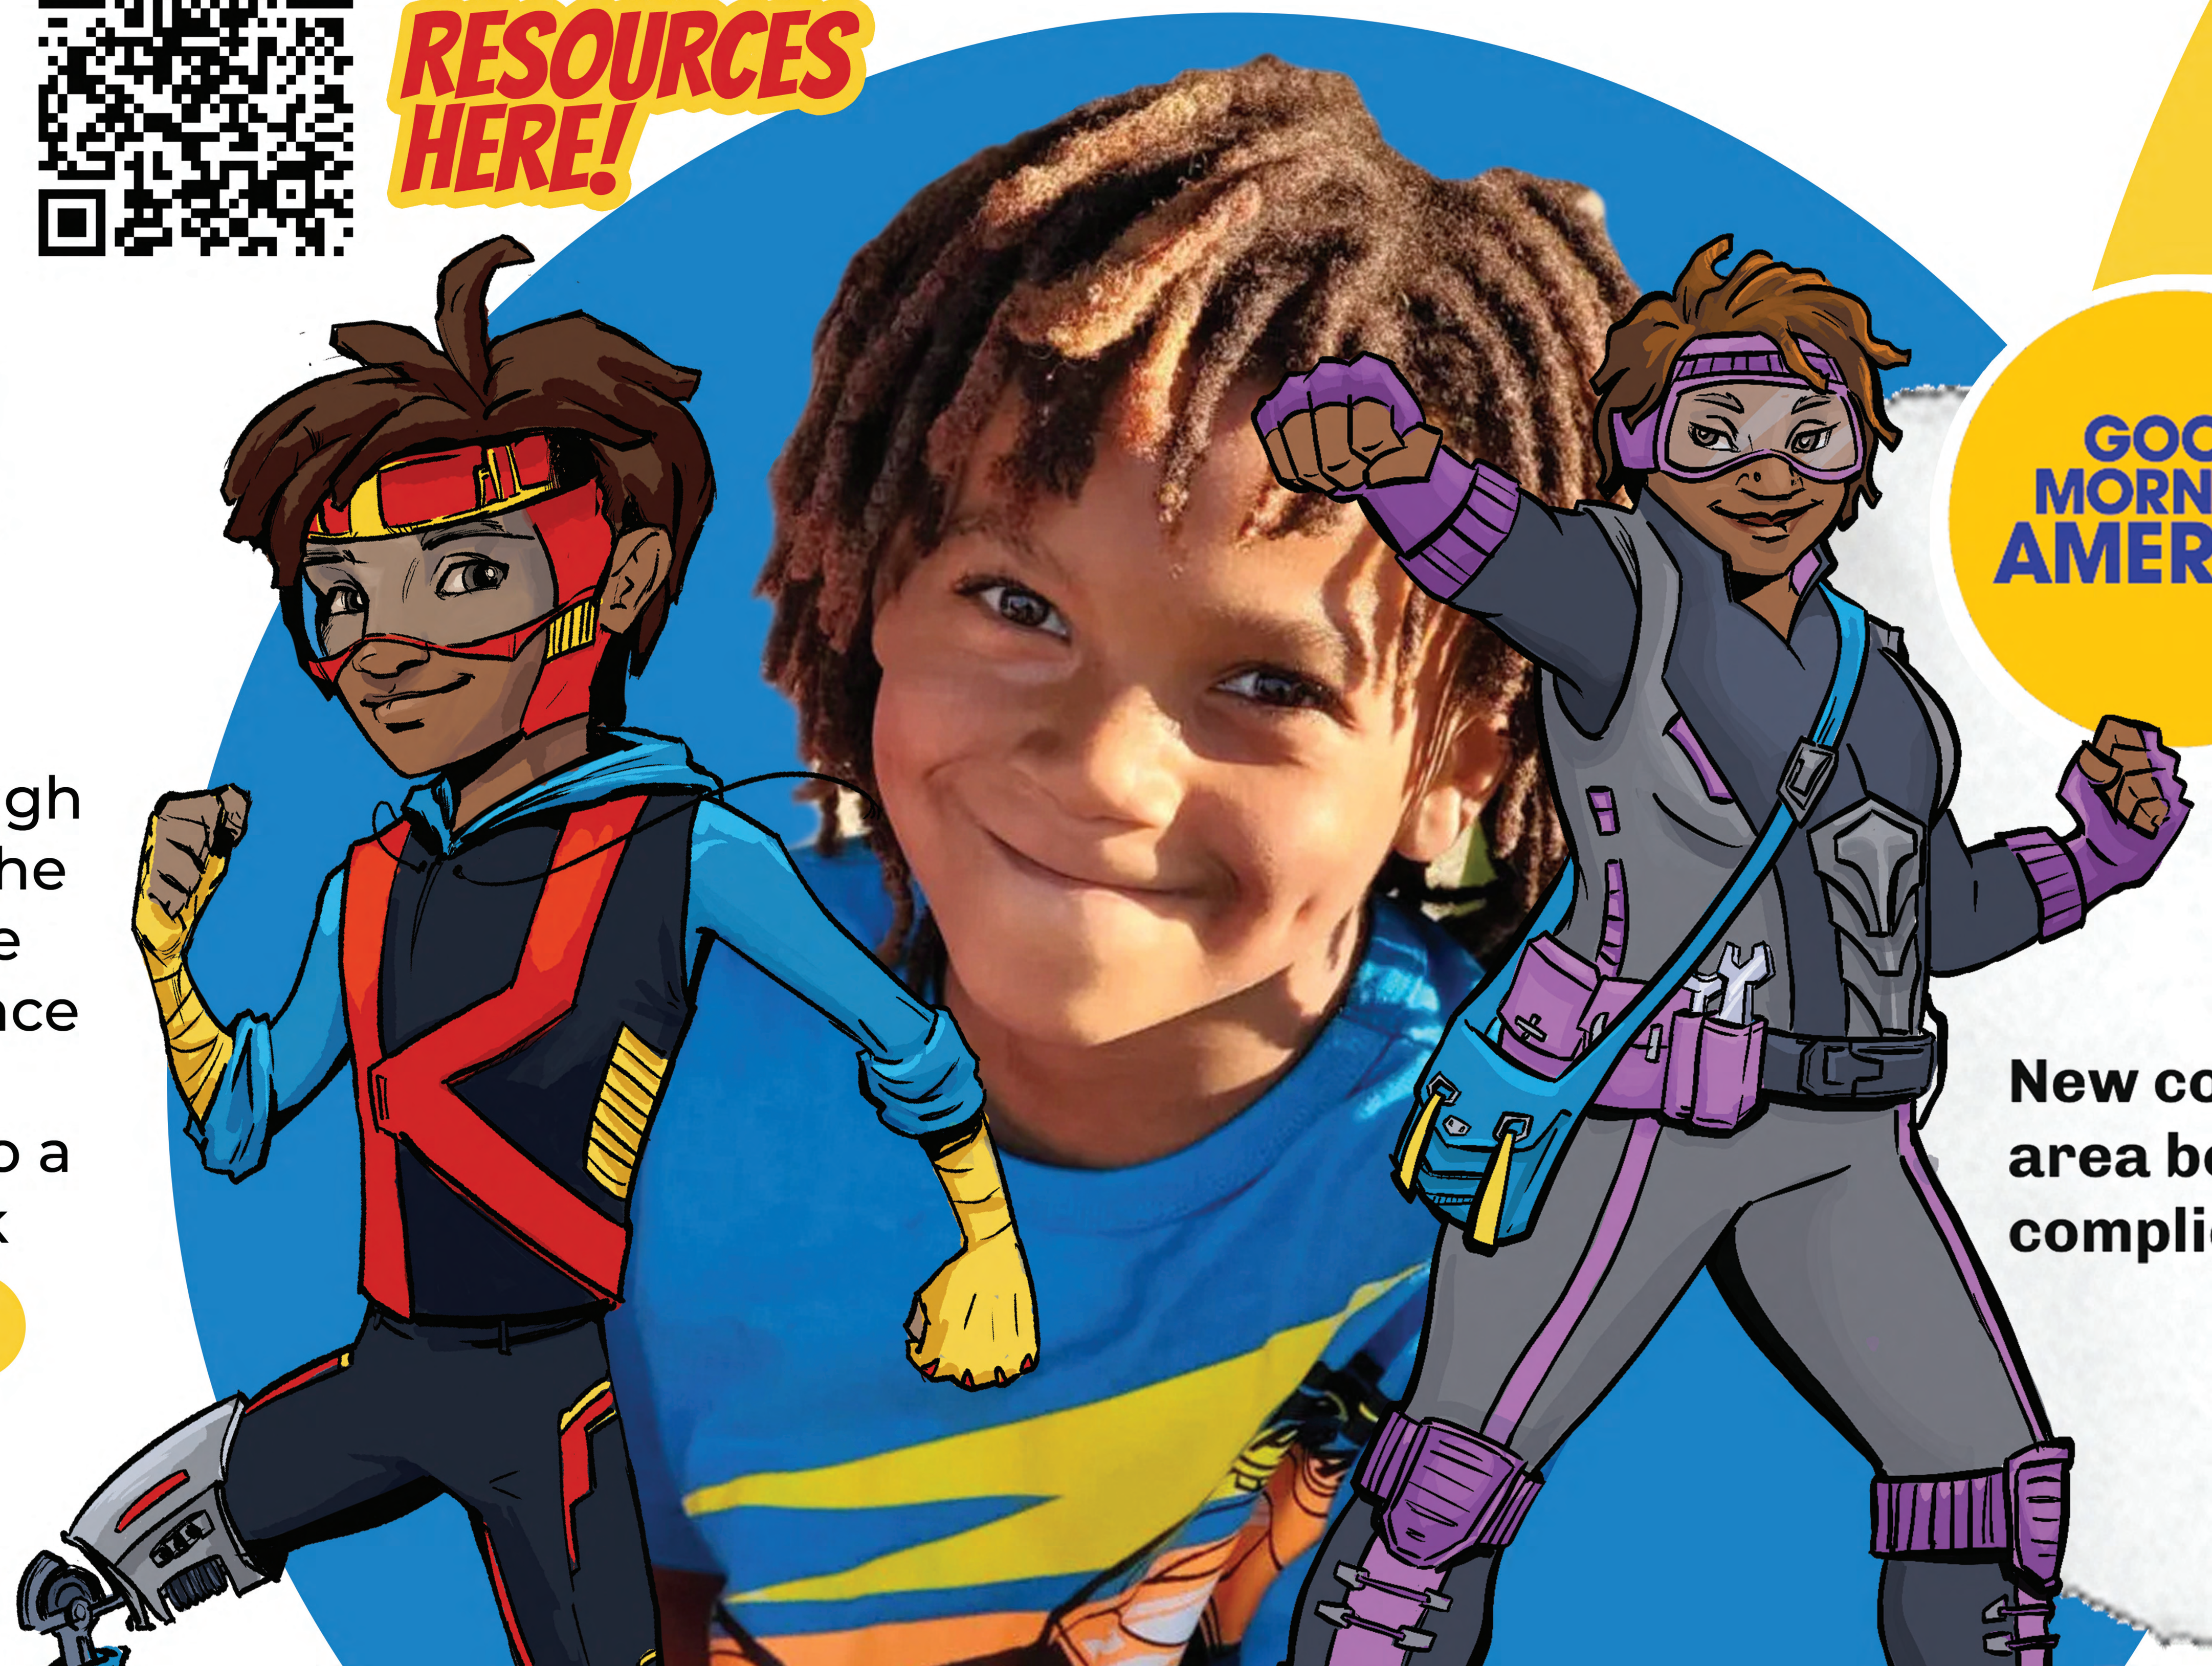

SUCCESS!

330+ million reached

through earned media and social media engagement

1,000+ comic books distributed

demonstrating how elevating real family stories into innovative tools inspires **awareness**, **empathy**, and **action** to fight the flu.

GOOD MORNING AMERICA

7-Year-Old Boy Who Lost Legs After Complications from the Flu Urges Everyone to Get the Flu Vaccine

Kaden Stevenson went into toxic shock syndrome from the flu, causing his heart, liver and kidneys to fail, resulting in his legs having to be amputated

By Charna Flam and Greta Bjornson | Updated on December 4, 2025 06:37PM EST

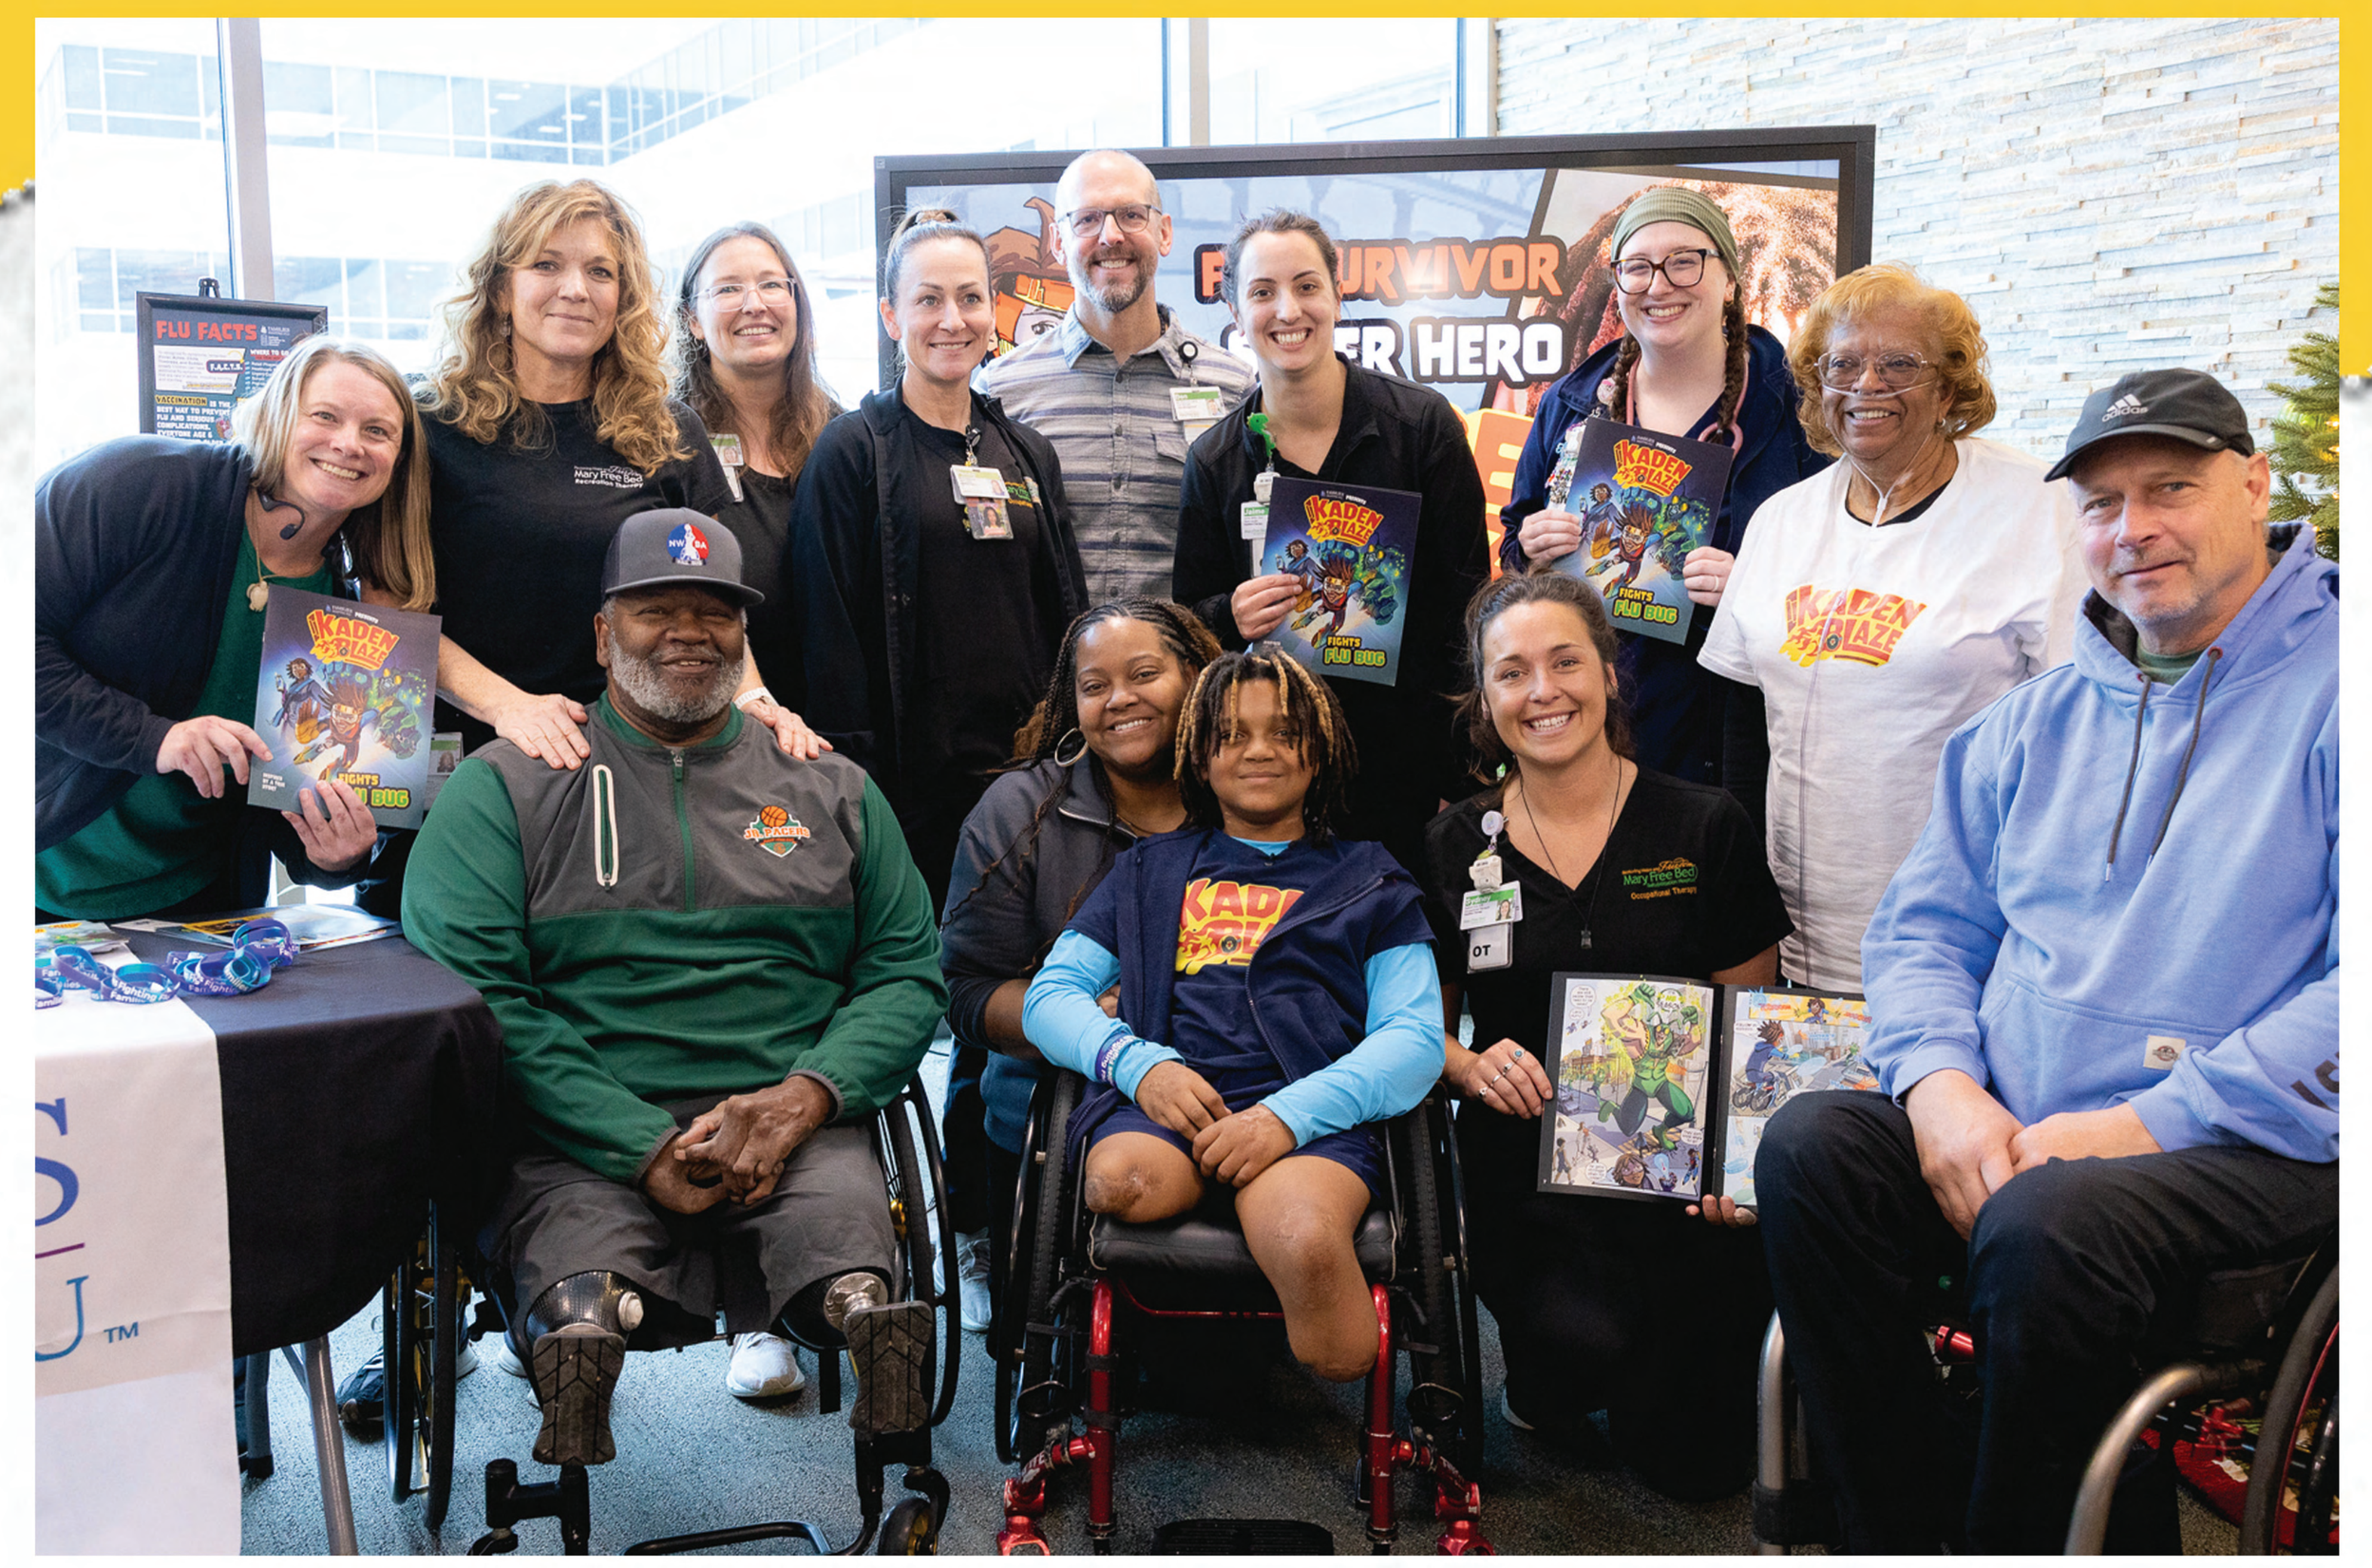

People

NBC NEWS

LOCAL 4 CLICK ON DETROIT

New comic book based on the true story of an area boy who lives with double amputation after complications from the flu

abc 12 NEWS

THE FLINT Courier NEWS

WNEM 5

Boy's flu battle inspires new comic book, Flint Council elects president and other Flint-area news

Published: Sep. 28, 2025, 8:00 a.m.

MID-MICHIGAN NOW 25 FOX66