Poster #15

Availability of Live Attenuated Influenza Vaccine for Self- or Caregiver-Administration at Home: Description of the Patient Journey

Monisha Prakash,[1] Ami Patel,[2] Allyn Bandell[1]

[1]US Medical Affairs, AstraZeneca, Wilmington, DE, USA;
[2]Hub Operations, ASPN Pharmacies, LLC, NJ, USA

Scan here or follow link for e-poster*

Introduction

- Influenza vaccination is the most effective way to prevent severe disease caused by influenza infection;[1,2] however, in the United States (US), influenza vaccination coverage in adults and children remains below target levels.[3]
 - In addition, disparities in coverage by race and ethnicity persist in the US. These disparities have worsened among people living in rural areas compared with those living in suburban or urban areas.[4]
- Live attenuated influenza vaccine (LAIV, FLUMIST®) is a needle-free influenza vaccine given as a nasal spray,[5] and was recently approved by the US Food and Drug Administration for self- or caregiver- administration.[6]
- Here, we describe the LAIV self-administration ordering process via a computer or smartphone-optimized website.

Conclusions

- The digital LAIV patient journey resembles other medical-related online processes, and is similar to registration for an in-person immunization appointment at a pharmacy.
- The convenience and flexibility of LAIV self- or caregiver-administration may be optimal for some individuals and families and improves influenza vaccine access in the US.

*E-poster Copies of this poster obtained through the QR code or link are for personal use only and may not be reproduced without permission.

Presented at the National Adult and Influenza Immunization Summit Meeting 2025 | May 13–15, 2025 | Atlanta, GA, USA

LAIV Patient Journey: Ordering, Shipping, and At-Home Administration

1 Finding out & learning more about FLUMIST

FLUMIST AstraZeneca website

Includes educational resources & support[7]

FLUMIST ordering website

APSN pharmacy website that supports FLUMIST home service

Adult enters their information to **initiate order.**

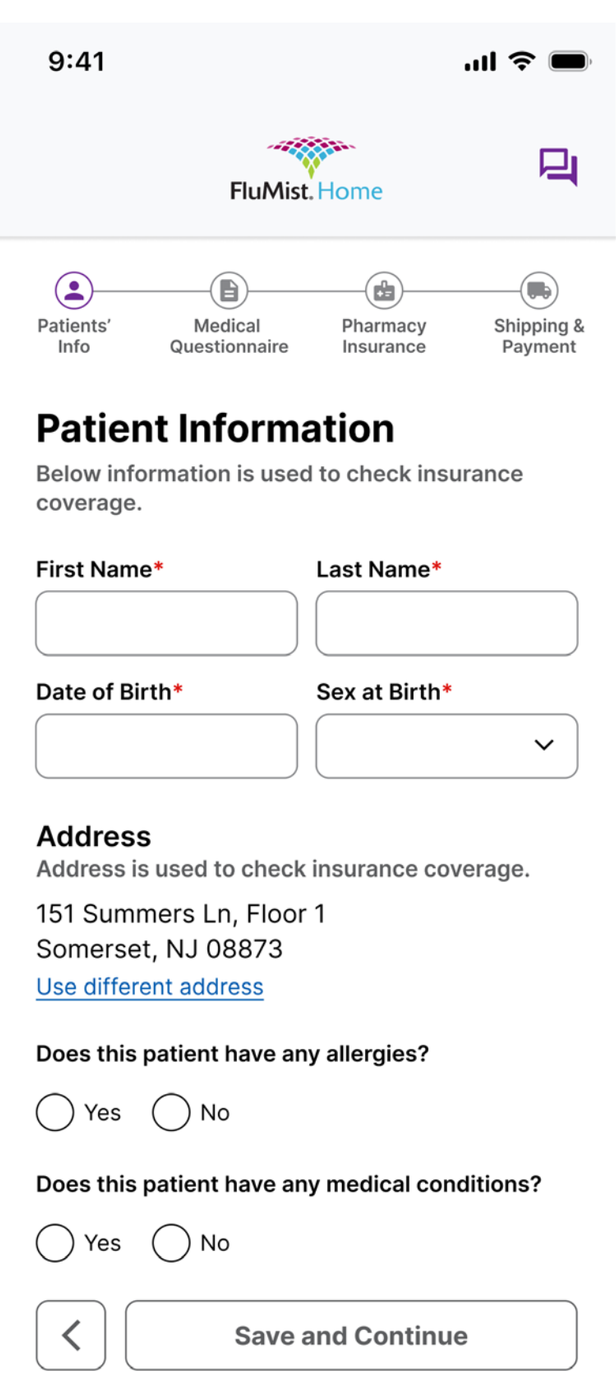

9:41
FluMist.Home
Patients' Info
Medical Questionnaire
Pharmacy Insurance
Shipping & Payment
Patient Information
Below information is used to check insurance coverage.
First Name*
Last Name*
Date of Birth*
Sex at Birth*
Address
Address is used to check insurance coverage.
151 Summers Ln, Floor 1
Somerset, NJ 08873
Use different address
Does this patient have any allergies?
Yes No
Does this patient have any medical conditions?
Yes No
Save and Continue

2 Ordering through ASPN pharmacies, LLC[a]

Adult completes an online medical screening questionnaire to **determine eligibility**

consistent with ACIP guidance.

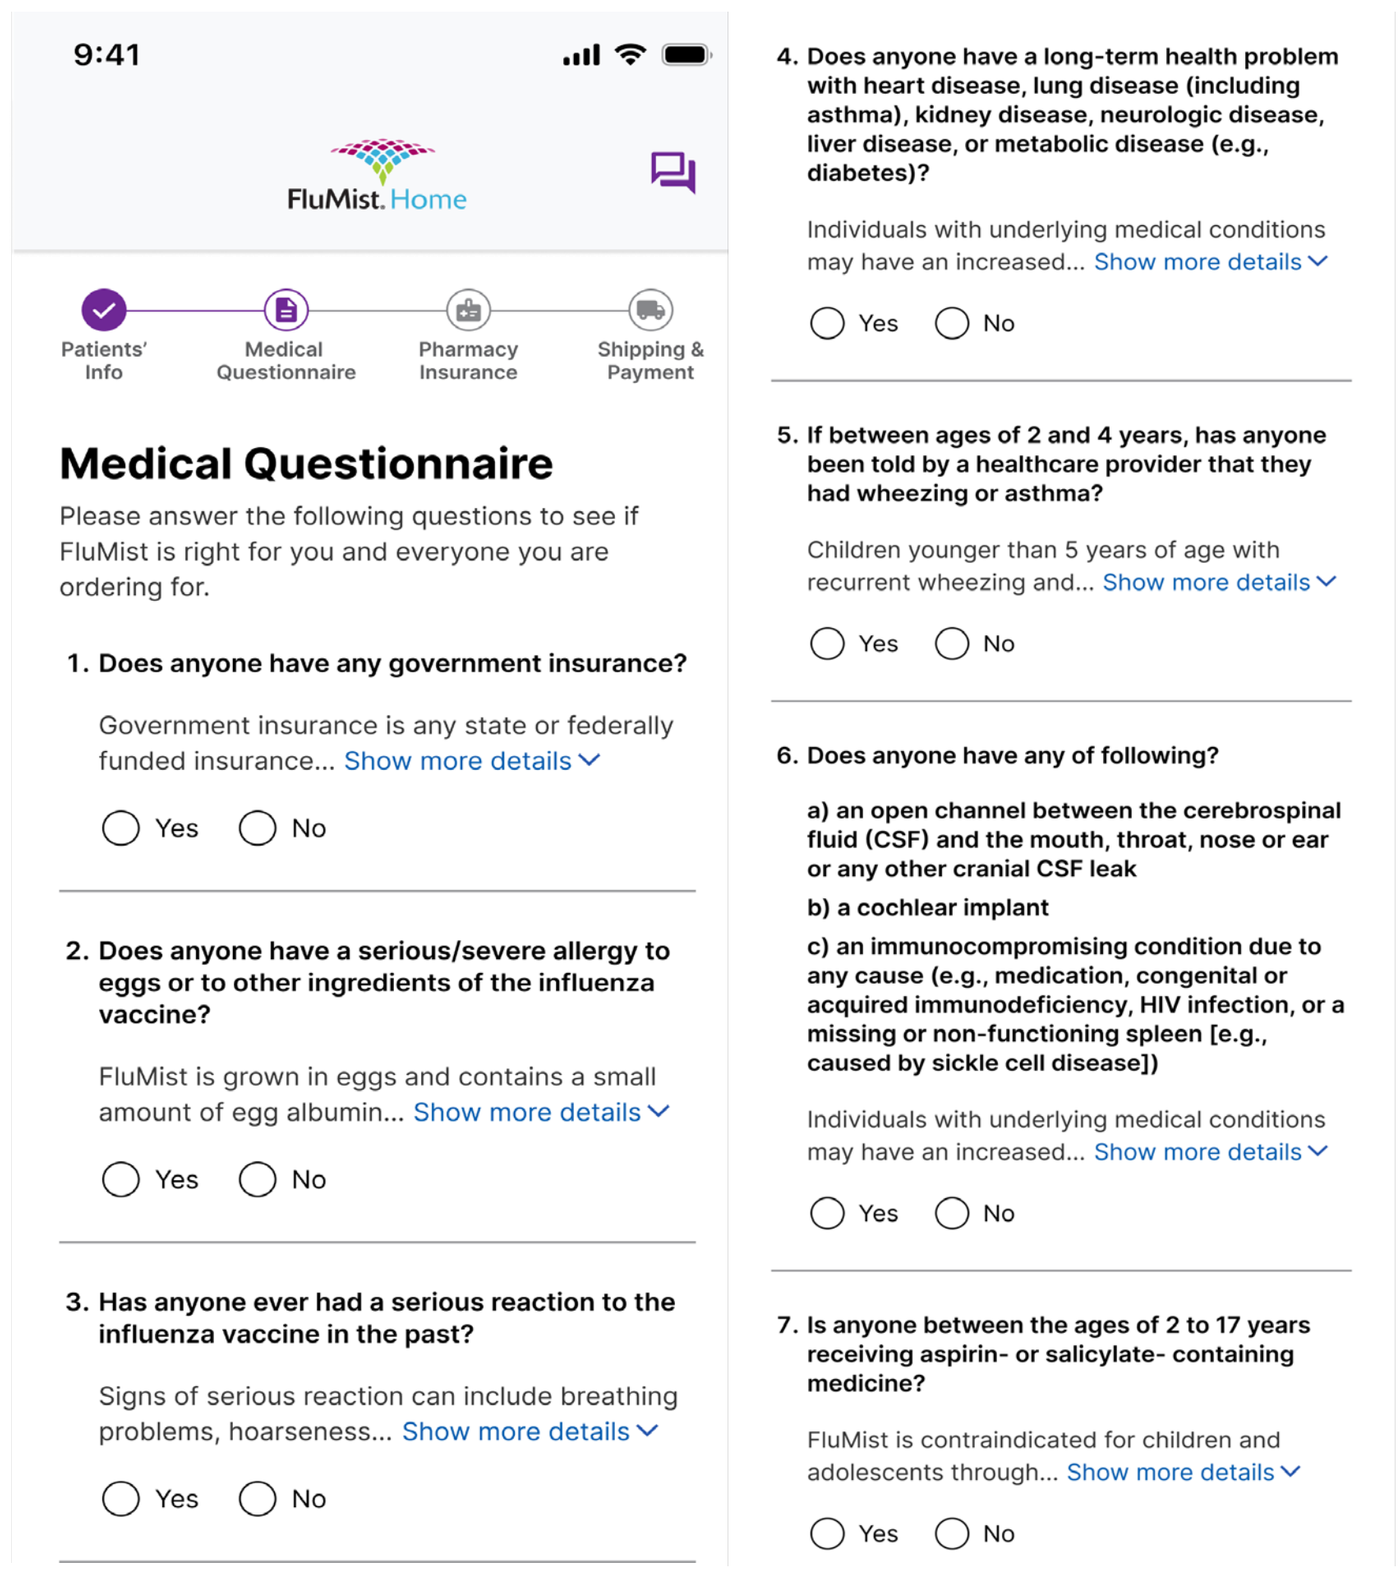

9:41
FluMist.Home
Patients' Info
Medical Questionnaire
Pharmacy Insurance
Shipping & Payment
Medical Questionnaire
Please answer the following questions to see if FluMist is right for you and everyone you are ordering for.
1. Does anyone have any government insurance?
Government insurance is any state or federally funded insurance... Show more details
Yes No
2. Does anyone have a serious/severe allergy to eggs or to other ingredients of the influenza vaccine?
FluMist is grown in eggs and contains a small amount of egg albumin... Show more details
Yes No
3. Has anyone ever had a serious reaction to the influenza vaccine in the past?
Signs of serious reaction can include breathing problems, hoarseness... Show more details
Yes No
4. Does anyone have a long-term health problem with heart disease, lung disease (including asthma), kidney disease, neurologic disease, liver disease, or metabolic disease (e.g., diabetes)?
Individuals with underlying medical conditions may have an increased... Show more details
Yes No
5. If between ages of 2 and 4 years, has anyone been told by a healthcare provider that they had wheezing or asthma?
Children younger than 5 years of age with recurrent wheezing and... Show more details
Yes No
6. Does anyone have any of following?
a) an open channel between the cerebrospinal fluid (CSF) and the mouth, throat, nose or ear or any other cranial CSF leak
b) a cochlear implant
c) an immunocompromising condition due to any cause (e.g., medication, congenital or acquired immunodeficiency, HIV infection, or a missing or non-functioning spleen [e.g., caused by sickle cell disease])
Individuals with underlying medical conditions may have an increased... Show more details
Yes No
7. Is anyone between the ages of 2 to 17 years receiving aspirin- or salicylate- containing medicine?
FluMist is contraindicated for children and adolescents through... Show more details
Yes No

These questions are representative of the medical questionnaire.

[a]The online pharmacy for self- or caregiver-administered FLUMIST.

3 Confirming eligibility

HCP reviews the medical questionnaire, **confirms eligibility,** and generates and fills prescription[b]

per state-specific pharmacy regulations.

- **Commercial or TRICARE insurance for the 2025/26 season**
 Only private/TRICARE health insurance
- **Age[8]**
 2–49 years
- **Medical eligibility**
 ACIP guidance

- **Pharmacist is available** via chat or **toll-free number** for questions
- Eligibility determined per Immunize.org **LAIV screening tool.[8]**

[b]Based on patient shipping address.

4 Shipping & delivery

Shipped under **strict cold chain logistics** using validated packaging and delivery methods which were part of the FDA approval.

Patients will receive **tracking updates** and/or **delivery notifications** via text/email.

iMessage
Today 2:16 PM
Your FluMist Rx has been sent to [pharmacy name]. You will receive a tracking number as soon as it is available.

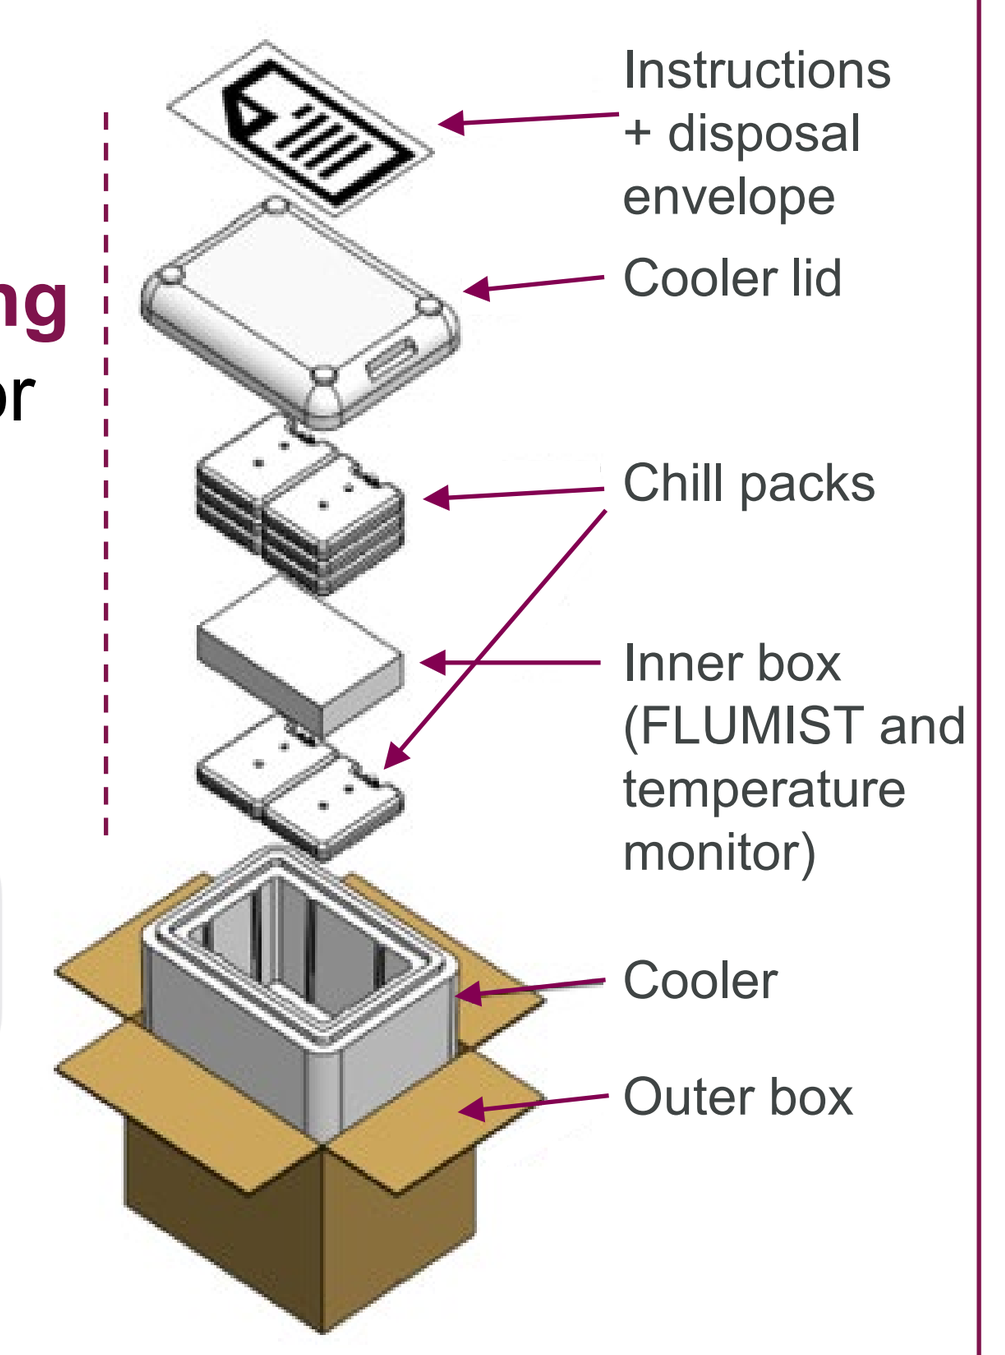

Instructions + disposal envelope
Cooler lid
Chill packs
Inner box (FLUMIST and temperature monitor)
Cooler
Outer box

FLUMIST is delivered on the date selected by the patient.
No signature is required.

5 Handling & administration

Instructions for home **storage & administration** are provided via text and in shipping box

FLUMIST vaccine information statement is provided online, via email, and in shipping box

Additional resources:

- **QR code** for "How to administer FLUMIST" video
- **Online and toll-free number** for pharmacist support

Text messages are sent for adult to **confirm administration.**

iMessage
Today 2:16 PM
Record your Flumist dose by clicking here [URL] and download your vaccination record.

6 Disposal of FLUMIST sprayer & packaging

The used FLUMIST sprayer is considered medical waste. To dispose of appropriately, a **prepaid and prelabeled envelope is provided in the shipment. All used FLUMIST sprayers can be placed into the mailbox.**

Packing materials can be **reused, recycled or disposed** per local and federal guidelines

Reporting vaccination status through immunization information systems (IIS)[c]

Adult confirms administration of the vaccine. Pharmacy then reports vaccination to IIS.[9,10]

Adult > Confirms administration through online pharmacy website > Reporting service > Registries > Centers for Disease Control and Prevention dashboard

Documentation is available to patients and their health care providers.[d]

[c]In compliance with state requirements for consent and data components.
[d]If contact information provided.

Please note that the images of the processes shown above are examples of what the processes will look like and may change.

Acknowledgments

Medical writing support was provided by India Wright, MSc, and editorial support was provided by Isabella Cannava, BA, both of Core (a division of Prime, London, UK), and supported by AstraZeneca according to Good Publication Practice guidelines.

Disclosures

M.P and A.B are employees of, and stockholders in, AstraZeneca. A.P is an employee of ASPN Pharmacies, LLC.

References

1. World Health Organization. Available at: https://www.who.int/europe/news-room/fact-sheets/item/influenza-vaccination-coverage-and-effectiveness [Accessed April 02, 2025].
2. Centers for Disease Control and Prevention. Available at: https://www.cdc.gov/flu/prevention/index.html [Accessed April 02, 2025].
3. U.S. Department of Health and Human Services. Available at: https://odphp.health.gov/healthypeople/objectives-and-data/browse-objectives/vaccination/increase-proportion-people-who-get-flu-vaccine-every-year-iid-09 [Accessed April 02, 2025].
4. Centers for Disease Control and Prevention. Available at: https://www.cdc.gov/flu/whats-new/2023-2024-flu-coverage-update.html?CDC_AAref_Val=https://www.cdc.gov/flu/spotlights/2023-2024/flu-coverage-update.htm [Accessed April 02, 2025].
5. U.S. Food and Drug Administration. Available at: https://www.azpicentral.com/pi.html?product=flumist&country=us&popup=no [Accessed April 02, 2025].
6. U.S. Food and Drug Administration. Available at: https://www.fda.gov/news-events/press-announcements/fda-approves-nasal-spray-influenza-vaccine-self-or-caregiver-administration [Accessed April 02, 2025].
7. FluMist®. Available at: https://www.flumist.com/ [Accessed April 02, 2025].
8. Immunize.org. Available at: https://www.immunize.org/wp-content/uploads/catg.d/p4067.pdf [Accessed April 02, 2025].
9. Centers for Disease Control and Prevention. Available at: https://www.cdc.gov/iis/about/index.html#cdc_data_surveillance [Accessed April 02. 2025].
10. Centers for Disease Control and Prevention. Available at: https://www.cdc.gov/fluvaxview/dashboard/vaccine-administration-coverage-jurisdiction-iis.html [Accessed April 02, 2025].

ACIP = Advisory Committee on Immunization Practices.